P-1452

A Seasonal Influenza Hemagglutinin mRNA-Based Vaccine Induces Humoral and Cellular Immunity Comparable to an Adjuvanted Inactivated Influenza Virus Vaccine

Carole Henry, Daniel Makrinos, Runxia Liu, Maria Cavallaro, Brooke Fenderson, Yanbo Sun, Eleanor Astley, Bethany Girard, Wen-Han Yu, Anthony DiPiazza, Jaap Oostendorp,* Robert Paris

Moderna, Inc., Cambridge, MA, USA
*Presenting author.

325 Binney Street
Cambridge, MA 02142, USA
Jaap.Oostendorp@modernatx.com

BACKGROUND

- Seasonal influenza continues to cause substantial morbidity and mortality worldwide[1]
- Traditional influenza vaccines are manufactured in eggs; mutations may occur that alter antigenicity and reduce efficacy[2,3]
- Alternative platforms, including recombinant hemagglutinin (HA) vaccines and subunit vaccines from mammalian cells, have been developed to address limitations of the egg-based vaccines[4,5]
- Adjuvanted vaccines, such as FLUAD with MF59, enhance immune responses and broaden protection[6]
- mRNA vaccines offer strong immunogenicity, the flexibility to incorporate multiple antigens, and the capacity for rapid update[7]
- mRNA-1010, an mRNA-based seasonal influenza HA vaccine, elicited superior immune responses relative to licensed standard- or high-dose seasonal egg-based influenza vaccines in adults aged ≥18 years in a recent phase 3 trial[8]; however, no direct comparisons of immune responses induced by mRNA-1010 versus adjuvanted vaccines have been conducted

OBJECTIVE

- To compare the humoral and cellular immune responses induced by mRNA-1010 with those of a licensed adjuvanted egg-based influenza virus vaccine (FLUAD) across sex- and age-matched individuals in healthy adults 18 to 75 years of age

METHODS

Study Design

- This randomized, open-label, phase 1 trial evaluated the immunogenicity of mRNA-1010 (seasonal influenza quadrivalent HA mRNA vaccine) compared with FLUAD (adjuvanted influenza virus vaccine) in adults 18 to 75 years of age
- Participants were randomly assigned 1:1 to receive a single dose of mRNA-1010 (50 µg) or FLUAD (60 µg), with an approximately equal distribution of age groups (18-49 and 50-75 years; **Figure 1**)
- For both vaccine compositions, the 2022-2023 seasonal influenza World Health Organization recommendation for cell- (mRNA-1010) and egg-based (FLUAD) quadrivalent vaccines were followed
- Blood samples were collected from vaccinated individuals at baseline (Day 1), 7 days (Day 8), 14 days (Day 15), 28 days (Day 29), 90 days (Day 91), 180 days (Day 181), and 360 days (Day 361) following vaccination as part of the mRNA-CRID-001 clinical trial (NCT05397223)
- This analysis presents the humoral and cellular immune responses induced by mRNA-1010 against 4 influenza strains: A/Wisconsin/588/2019 (A/H1N1), A/Darwin/6/2021 (A/H3N2), B/Austria/1359417/2021 (B/Victoria lineage), and B/Phuket/3073/2013 (B/Yamagata lineage)

Figure 1. Study Design

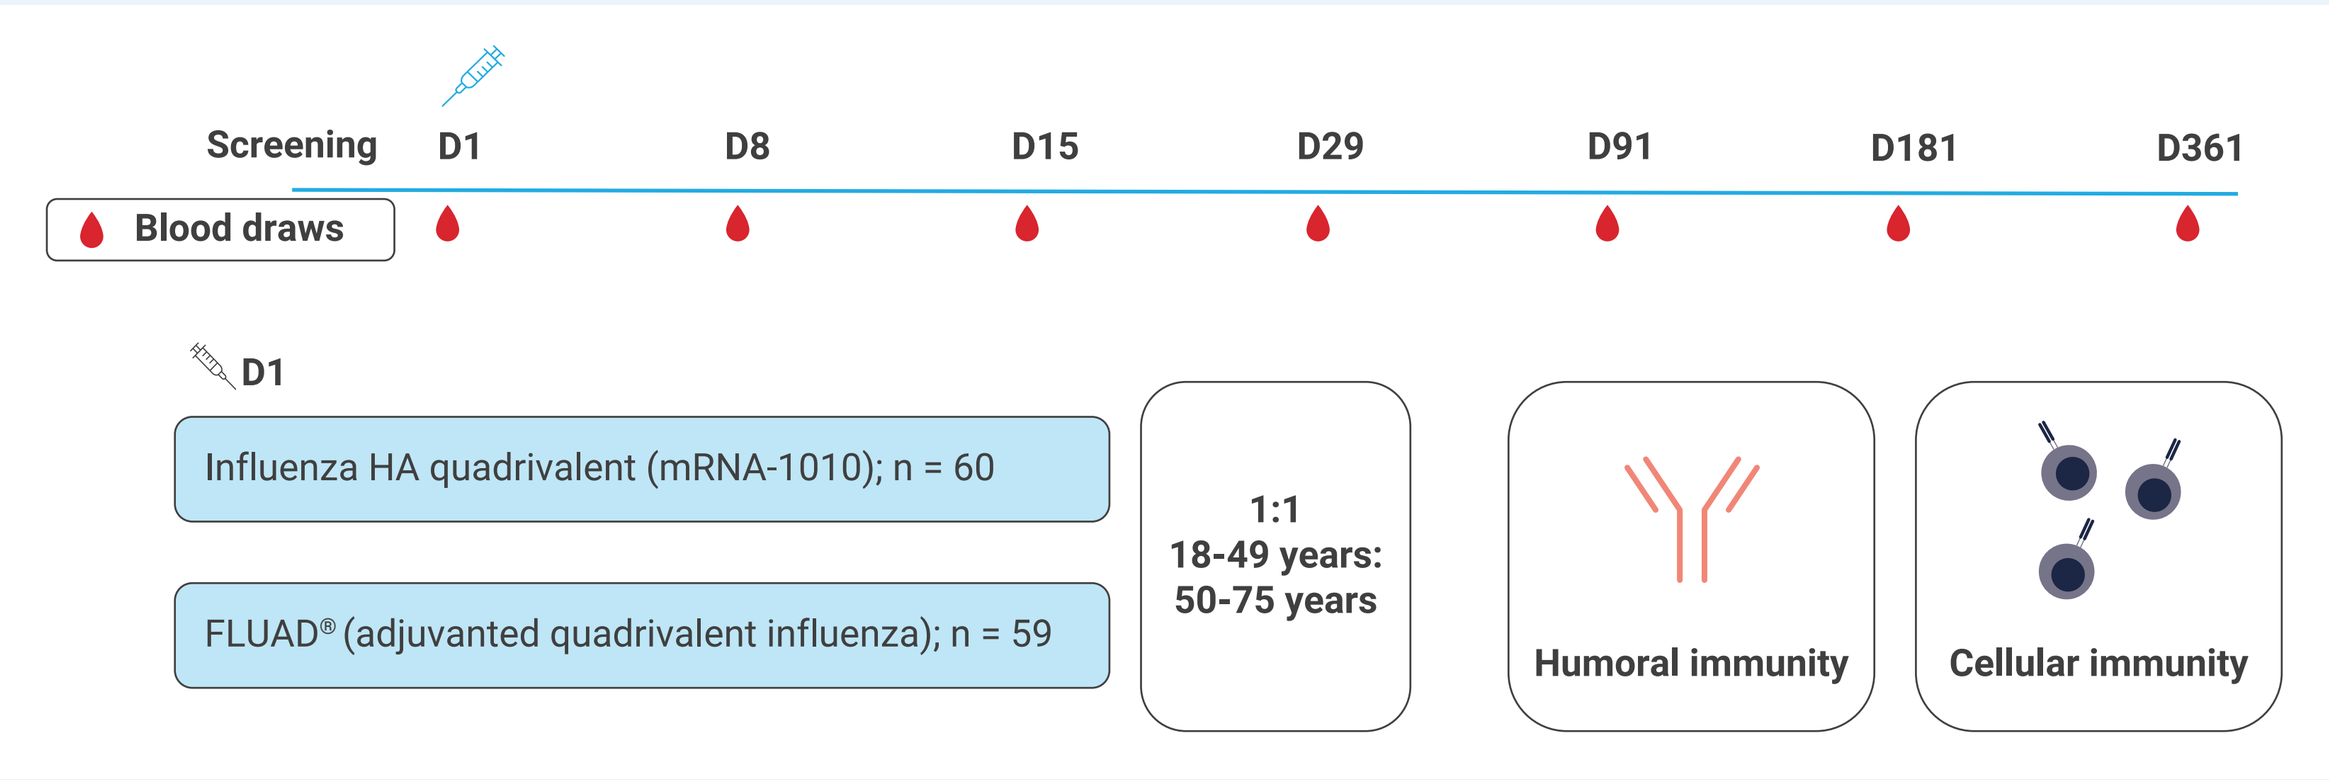

Screening
D1
D8
D15
D29
D91
D181
D361
Blood draws
D1
Influenza HA quadrivalent (mRNA-1010); n = 60
FLUAD® (adjuvanted quadrivalent influenza); n = 59
1:1
18-49 years:
50-75 years
Humoral immunity
Cellular immunity

D, day.

Assessment of Antibody Response, Antigen-Specific Memory B Cells, and Hemagglutinin-Specific T-Cell Responses

- Multiple sample types were collected at different time points for assessment of humoral and cellular immune responses to the 4 influenza strains, which were evaluated using hemagglutination inhibition (HAI) assays, flow cytometry–based memory B-cell (MBC) profiling, and intracellular cytokine staining for T-cell characterization

RESULTS

Participants

- Participants received a single dose of mRNA-1010 (n = 60) or FLUAD (n = 59)
- The mean age was 46 years in both groups
- In the mRNA-1010 and FLUAD groups, 53.3% and 44.1% were female, and 70.0% and 74.6% were White, respectively

HAI Responses

- Serum samples were collected on Days 1 (baseline), 8, 15, 29 (mRNA-1010: n = 55; FLUAD: n = 57), 91 (mRNA-1010: n = 52; FLUAD: n = 54), 181 (mRNA-1010: n = 49; FLUAD: n = 52), and 361 (mRNA-1010: n = 44; FLUAD: n = 45)
- Both mRNA-1010 and FLUAD elicited HAI responses that were comparable and durable up to 12 months after vaccination for all 4 strains assessed (**Figure 2**)
 - Overall, GMTs were similar after a single dose of mRNA-1010 or FLUAD at Days 91, 181, and 361 (**Figure 2A**)
 - While peak titers at Days 15 and 29 were generally higher in the younger cohort (18-49 years) compared with the older cohort (50-75 years), no differences were observed at Days 181 and 361 (**Figure 2B** and **2C**)

Figure 2. HAI Responses Elicited by mRNA-1010 and FLUAD in (A) All Participants, (B) Adults 18-49 Years of Age, and (C) Adults 50-75 Years of Age

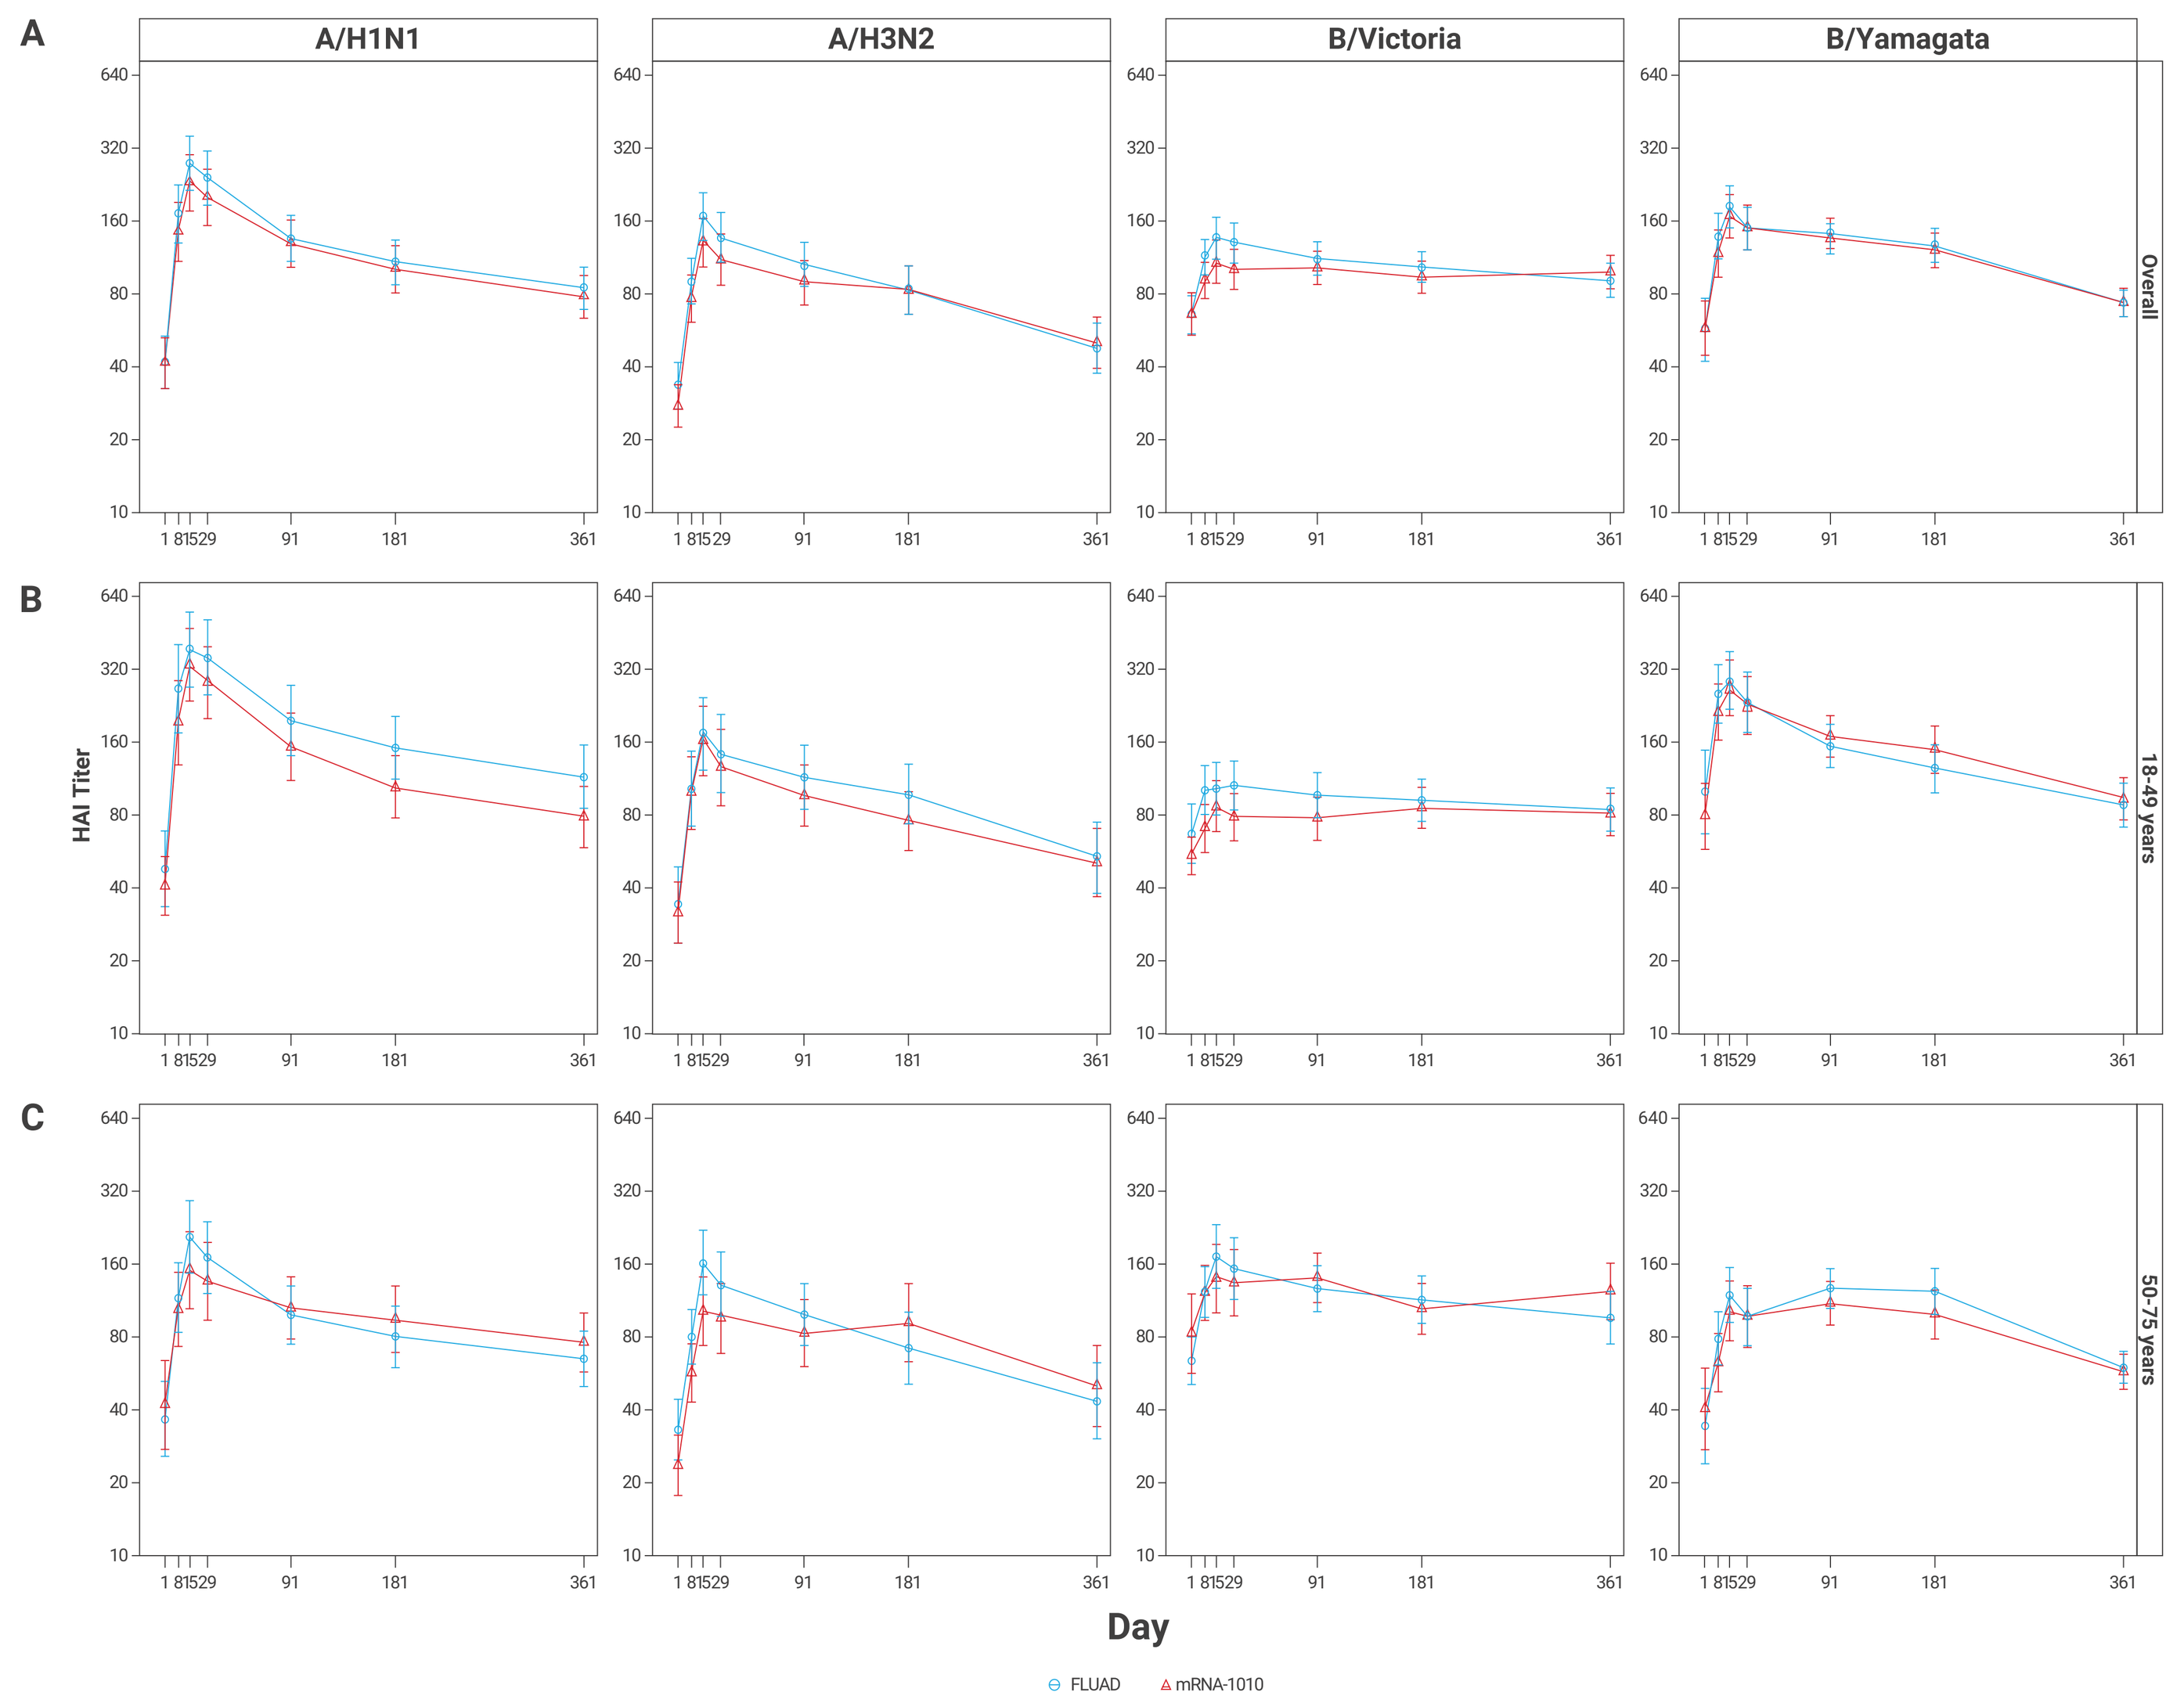

A
B
C
A/H1N1
A/H3N2
B/Victoria
B/Yamagata
Overall
18-49 years
50-75 years
HAI Titer
Day
FLUAD
mRNA-1010

HAI, hemagglutination inhibition; LLOQ, lower limit of quantification; ULOQ, upper limit of quantification.
Antibody values reported as below the LLOQ are replaced by 0.5 × LLOQ. Values greater than the ULOQ are converted to the ULOQ.
Influenza A H1N1 antibody (titer): LLOQ: 10, ULOQ: 1280 from Days 1-29, and LLOQ: 10, ULOQ: 4305 from Days 91-361; influenza A H3N2 antibody (titer): LLOQ: 10, ULOQ: 2560 from Days 1-29, and LLOQ: 10, ULOQ: 4305 from Days 91-361; influenza B/Victoria-lineage (titer): LLOQ: 10, ULOQ: 640 from Days 1-29, and LLOQ: 10, ULOQ: 4561 from Days 91-361; influenza B/Yamagata-lineage (titer): LLOQ: 10, ULOQ: 2560 from Days 1-29, and LLOQ: 10, ULOQ: 5120 from Days 91-361.

HA-Specific Memory B-Cell Responses

- Antigen-specific MBC responses were assessed in cryopreserved PBMC samples collected on Days 1 (mRNA-1010: n = 54; FLUAD: n = 56), 29 (mRNA-1010: n = 55; FLUAD: n = 55), and 91 (mRNA-1010: n = 54; FLUAD: n = 53)
- Robust HA-specific MBC responses were observed across all 4 influenza vaccine strains (**Figure 3**)
 - mRNA-1010 induced significantly higher frequencies of H3 A/Darwin/6/2021 HA-specific CD27⁺ MBCs at Days 29 and 91 compared with FLUAD (**Figure 3A**); the greatest response was observed in younger adults compared with older adults (**Figure 3B**)
 - mRNA-1010 induced significantly higher frequencies of activated CD27⁺CD21⁻ memory B cells specific to H3 when compared with FLUAD, while inducing similar MBC responses to the other 3 strains (**Figure 3C**)

Figure 3. HA-Specific Memory B-Cell Responses Induced by mRNA-1010 and FLUAD (A) at Days 1, 29, and 91 for All Participants, (B) at Day 29 Comparing Adults 18-49 Years of Age and 50-75 Years of Age, and (C) at Day 29 Comparing Classical CD27⁺CD21⁺ MBCs With Activated CD27⁺CD21^lo MBCs for All Participants

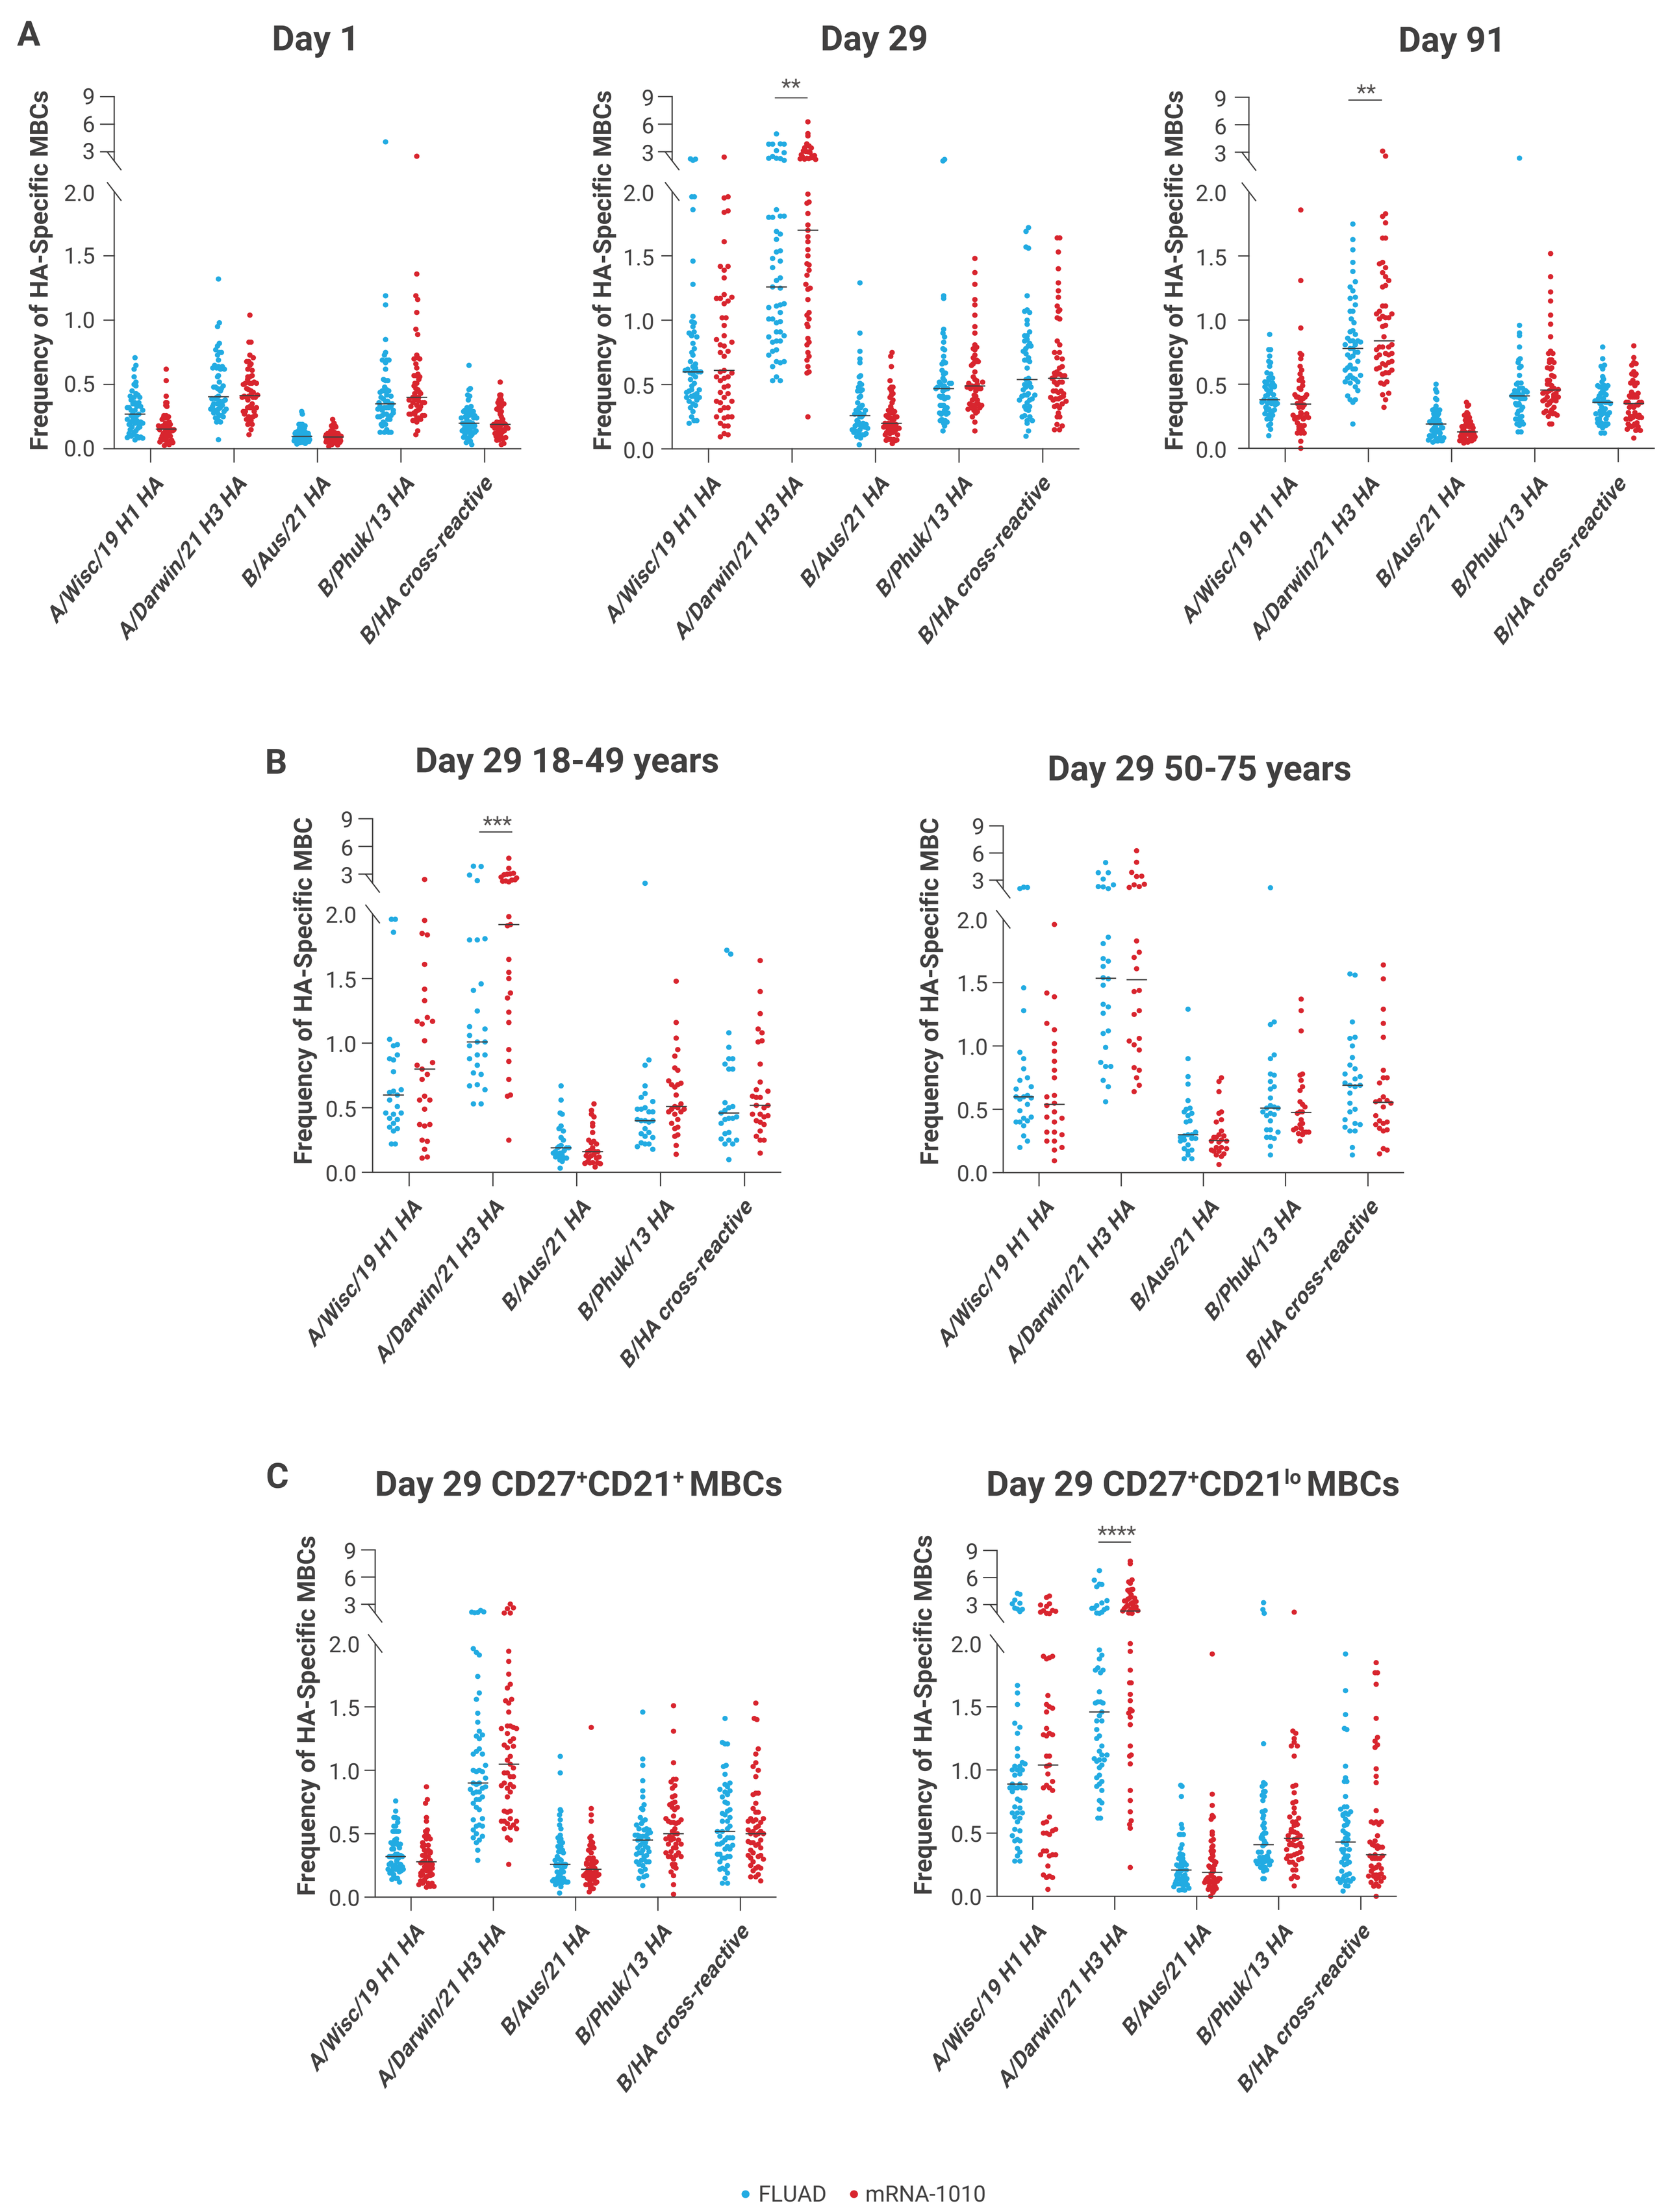

A
Day 1
Day 29
Day 91
Frequency of HA-Specific MBCs
A/Wisc/19 H1 HA
A/Darwin/21 H3 HA
B/Aus/21 HA
B/Phuk/13 HA
B/HA cross-reactive
B
Day 29 18-49 years
Day 29 50-75 years
Frequency of HA-Specific MBC
C
Day 29 CD27⁺CD21⁺ MBCs
Day 29 CD27⁺CD21^lo MBCs
FLUAD
mRNA-1010

ANOVA, analysis of variance; HA, hemagglutinin; MBC, memory B cell.
A two-way ANOVA test was used to test for differences between groups (*P<0.05, **P<0.005, ***P<0.0005, ****P<0.0001).

HA-Specific T-Cell Responses

- Antigen-specific T-cell responses were assessed in a subset of participants (n = 15 for mRNA-1010; n = 16 for FLUAD) on Days 1, 15, and 29
- mRNA-1010 and FLUAD generated strong and comparable HA-specific CD4⁺ T-cell responses (**Figure 4A**)
- A trend towards higher CD8⁺ T-cell responses for influenza B strains was observed in mRNA-1010 recipients compared with FLUAD recipients (**Figure 4B**)

Figure 4. HA-Specific (A) CD4⁺ and (B) CD8⁺ T-Cell Responses Boosted by mRNA-1010 and FLUAD

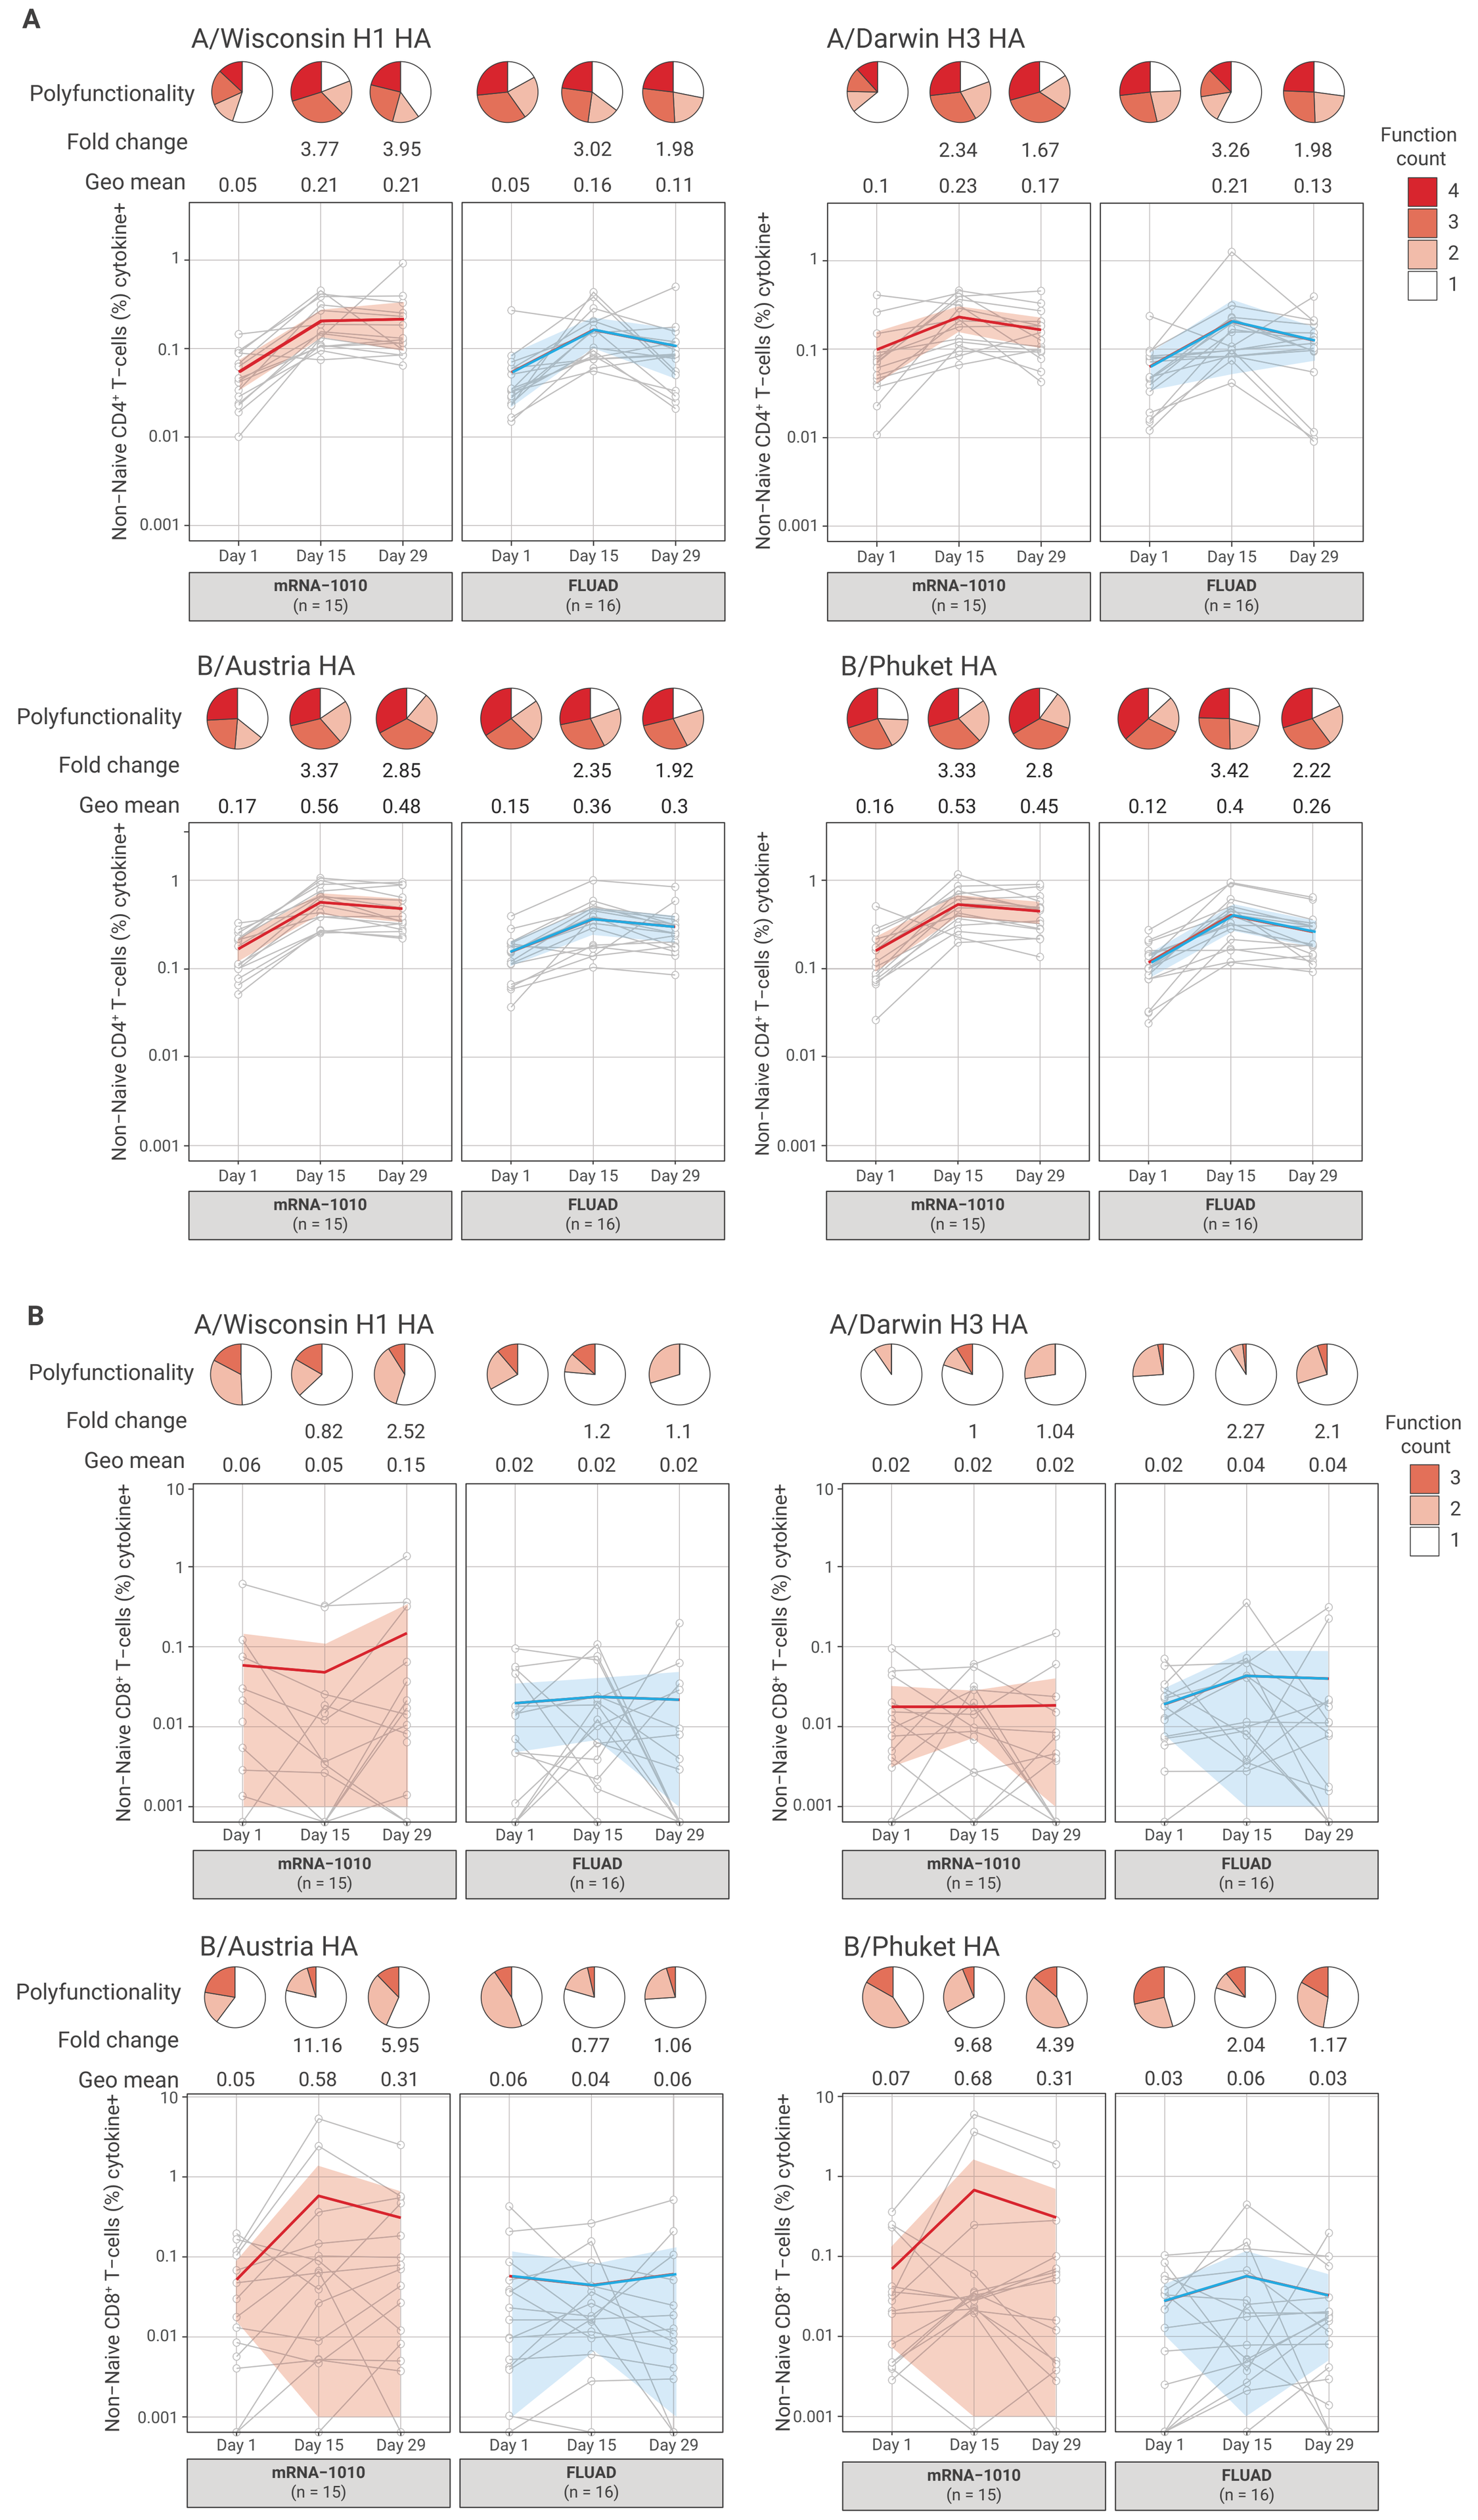

A
A/Wisconsin H1 HA
A/Darwin H3 HA
B/Austria HA
B/Phuket HA
Polyfunctionality
Fold change
Geo mean
Function count
Non−Naive CD4⁺ T−cells (%) cytokine+
Day 1
Day 15
Day 29
mRNA−1010 (n = 15)
FLUAD (n = 16)
B
Non−Naive CD8⁺ T−cells (%) cytokine+

ANOVA, analysis of variance; HA, hemagglutinin; IFN, interferon; IL, interleukin; TNF, tumor necrosis factor.
Antigen-specific CD8⁺ T cells are defined as CD8⁺ T cells positive for at least 1 of the following markers: IFN-γ, TNF-α, or IL-2. Each grey line represents an individual participant. The bold lines represent the geometric mean for each group: mRNA-1010 (in red) and FLUAD (in blue). The shaded areas around the bold lines indicate the 95% confidence intervals of the group means. The pie charts represent polyfunctionality, as the fraction of T cells expressing respective number of markers as indicated in the figure legend calculated from the arithmetic mean.
Significant differences between mRNA-1010 and FLUAD were observed using a two-way ANOVA with Šídák's multiple comparisons test at Days 15 and 29 for B/Austria (P=0.0203 and P=0.0438 respectively), and at Day 29 for B/Phuket (P=0.0298).

CONCLUSIONS

- These findings highlight the capacity of a first-generation mRNA-based influenza vaccine to induce robust humoral and cellular immunity, with a quality and breadth similar to that of an adjuvanted, enhanced seasonal influenza virus vaccine
- These data support the potential of the mRNA platform as a promising alternative to currently licensed seasonal influenza vaccines, with the additional advantages of the mRNA technology that can incorporate rapid strain updates and a flexible manufacturing platform that could enable the production and distribution of regionally matched influenza vaccines for improved effectiveness

ADDITIONAL INFORMATION

Please scan the QR code to access the digital poster and related conference materials. Copies of the poster obtained through this QR code are for personal use only and may not be reproduced without written permission of the authors.

For additional information, please contact Jaap Oostendorp (Jaap.Oostendorp@modernatx.com).

References

1. World Health Organization. *Wkly Epidemiol Rec*. 2022;97(19):185-208.
2. Chen H, et al. *Clin Infect Dis*. 2019;69(7):1198-1204. doi:10.1093/cid/ciy1065
3. Zost SJ, et al. *Proc Nat Acad Sci U S A*. 2017;114(47):12578-12583. doi:10.1073/pnas.1712377114
4. Cox MM, Hollister JR. *Biologicals*. 2009;37(3):182-189. doi:10.1016/j.biologicals.2009.02.014
5. Moro PL, et al. *Vaccine*. 2015;33(48):6684-6688. doi:10.1016/j.vaccine.2015.10.084
6. O'Hagan DT, et al. *Expert Rev Vaccines*. 2011;10(4):447-462. doi:10.1586/erv.11.23
7. Edwards DK, Carfi A. *Curr Opin Immunol*. 2022;77:102214. doi:10.1016/j.coi.2022.102214
8. Soens M, et al. *Vaccine*. 2025;50:126847. doi:10.1016/j.vaccine.2025.126847

Acknowledgments

The authors would like to acknowledge Guha Asthagiri Arunkumar for protein production and purification and Xiaolin Chang for Figure 2 visualization and statistical support. Medical writing and editorial assistance were provided by Renata Cunha, PharmD, of MEDiSTRAVA in accordance with Good Publication Practice guidelines, funded by Moderna, Inc., and under the direction of authors.

This study was funded by Moderna, Inc.

Disclosures

CH, DM, RL, MC, BF, YS, EA, BG, W-HY, ADP, JO, and RP are employees of Moderna, Inc., and may hold stock or stock options.

Presented at IDWeek 2025; October 19-22, 2025; Atlanta, GA, USA